HTD1801 (Berberine Ursodeoxycholate), A Unique Single Molecule with Multiple Beneficial Effects in Metabolic and Liver Diseases

Meng Yu, Ru Bai, Xinxiang Fu, Zhaobin Chen, Weijian Liu, Leigh MacConell

HighTide Therapeutics, Rockville, MD

BACKGROUND

- Multiple chronic liver diseases remain without adequate therapy and based on the complexity of pathophysiology require treatments with multiple mechanisms
- Ursodeoxycholic acid (UDCA) has beneficial effects in some liver diseases primarily through choleretic and anti-inflammatory effects
- Berberine (BBR) has a history of use in traditional medicine, emerging as a novel therapeutic with metabolic, antimicrobial, and anti-inflammatory effects
- Berberine ursodeoxycholate (HTD1801) is a new molecular entity - a salt of UDCA and BBR (1:1 stoichiometry), with benefits of its parent compounds and novel effects due to its unique molecular structure

Chemical Structure

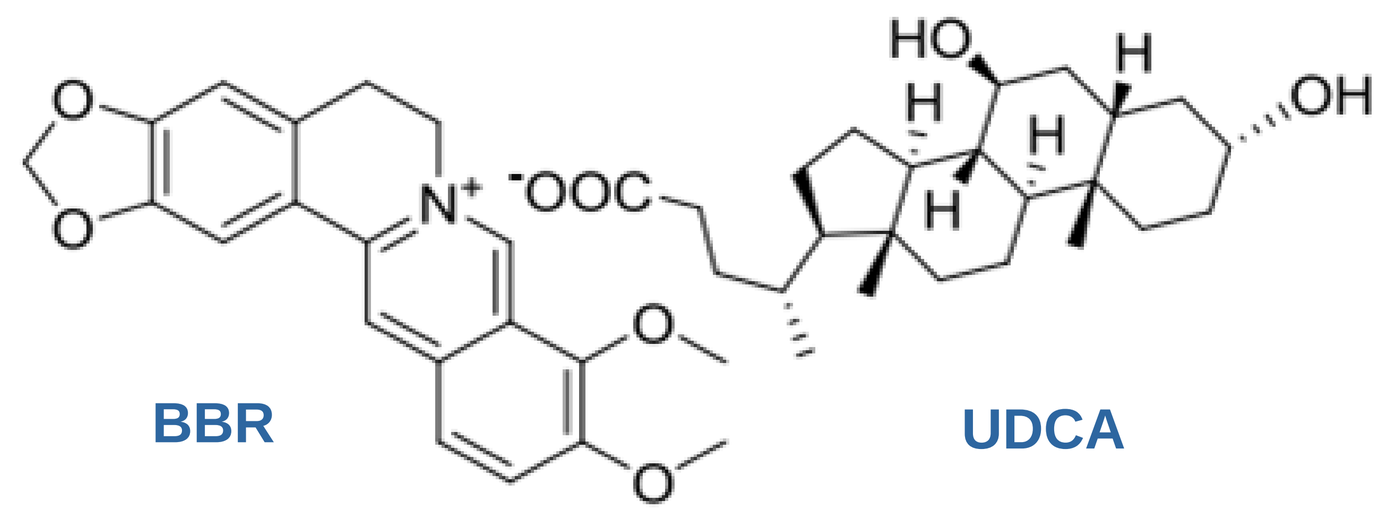
BBR
UDCA

- HTD1801 has been shown to be beneficial in human studies of hyperlipidemia, nonalcoholic steatohepatitis (NASH) and primary sclerosing cholangitis (PSC)

Aim

The purpose of this study was to demonstrate:

- Unique physicochemical characteristics of HTD1801
- Proof of concept of efficacy in a model of metabolic disease

METHODS

- The physicochemical properties of HTD1801, BBR chloride (BBR·Cl) and UDCA were compared via:
 - X-ray Powder Diffraction
 - Fourier Transform Infrared Spectroscopy (FTIR)
 - Melting Point Analysis
 - LogD Evaluation
 - Solubility Analysis (fasted and fed)
- Efficacy was evaluated in a preclinical NASH model (golden hamsters fed a high fat diet, 8/group) after 6 weeks of daily treatment of HTD1801, UDCA, or BBR·Cl

RESULTS

X-Ray Powder Diffraction, Fourier Infrared Spectroscopy, and Melting Point Analysis

X-Ray Powder Diffraction Patterns

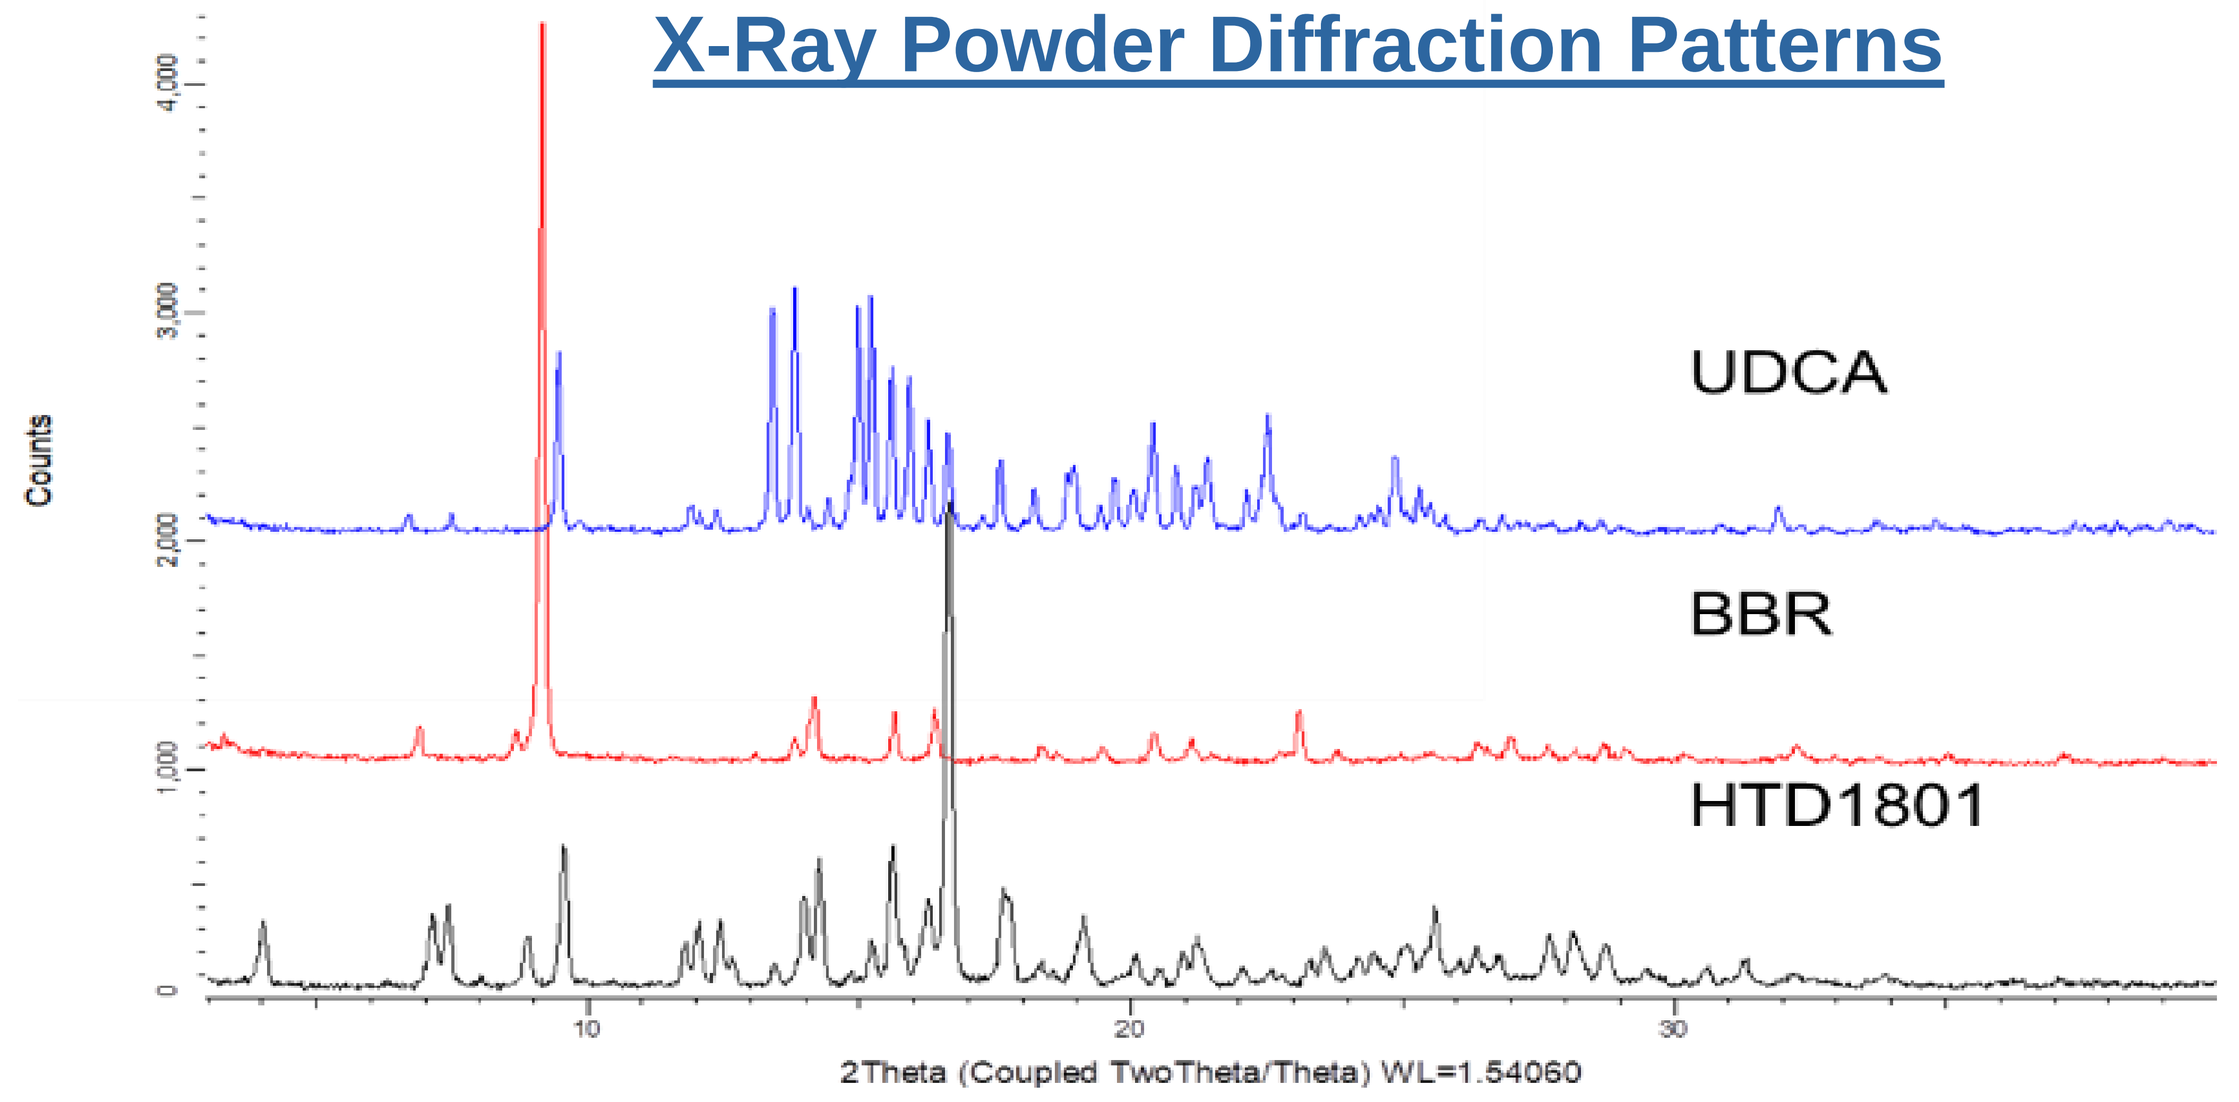
UDCA
BBR
HTD1801
Counts
2Theta (Coupled TwoTheta/Theta) WL=1.54060

- X-ray powder diffraction patterns and FTIR spectra support a unique crystal structure for HTD1801
- HTD1801 has a lower melting point (~120 ℃) vs BBR·Cl (204-206 ℃) and UDCA (200-205 ℃)
- The crystal form is a hemi-nonahydrate with a honeycomb-like 3D structure distinct from BBR·Cl and UDCA

Hemi-nonahydrate Single Crystal Structure

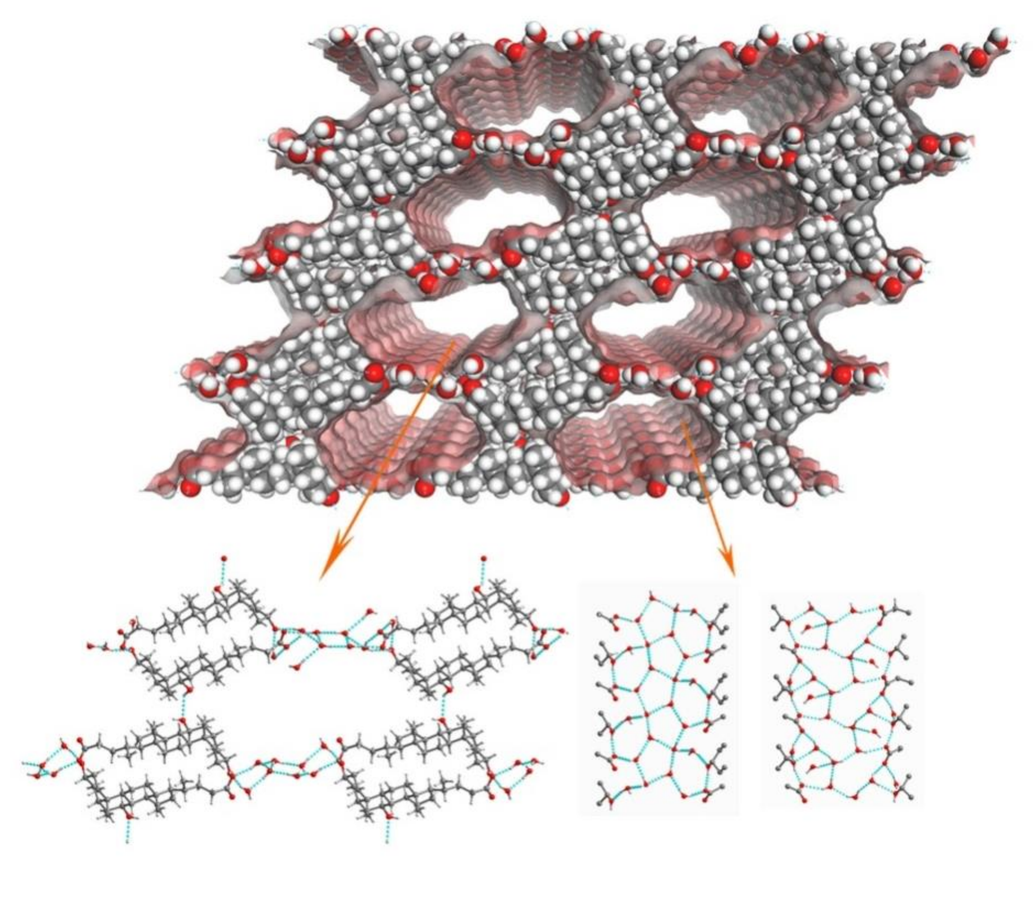

3D supramolecular structure with 1D linear channels of HTD1801

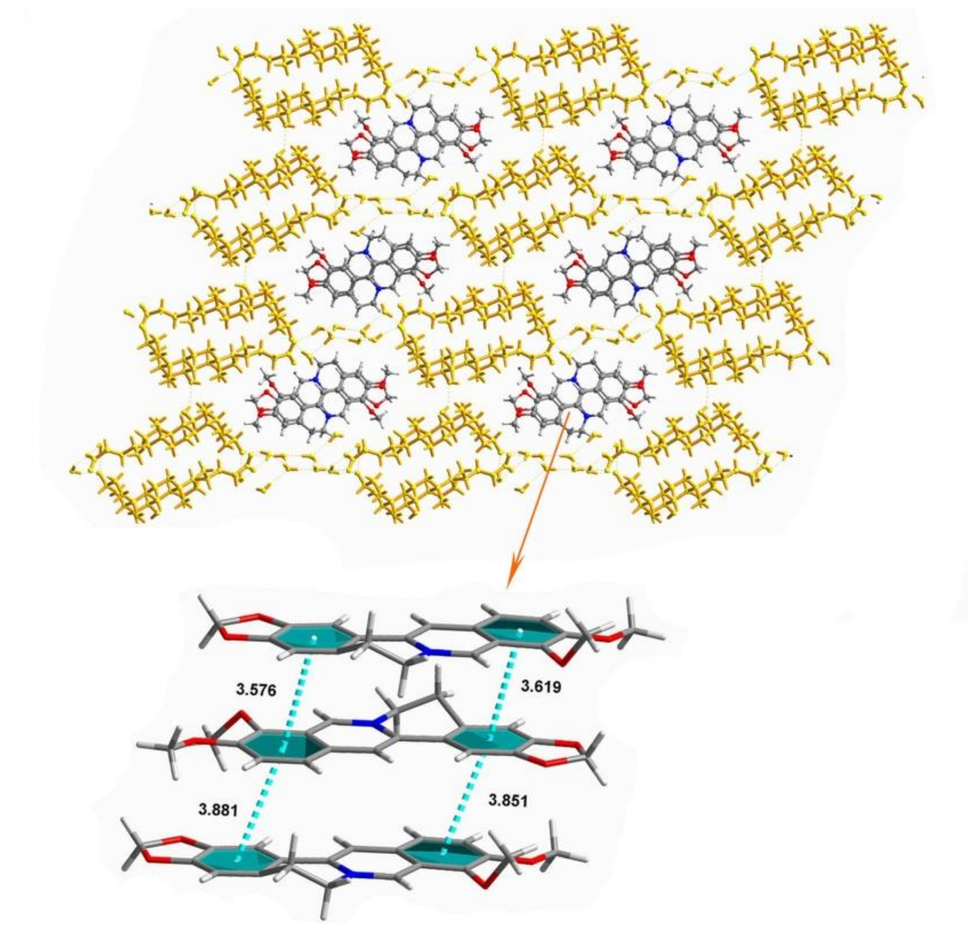

π-π stacking between BBR cations in HTD1801

Lipophilicity was Substantially Enhanced for BBR and UDCA as HTD1801

	LogD (Lipophilicity) Across the pH Range						
		2.0	**4.0**	**6.5**	**8.0**	**10.0**	**11.0**
BBR	**BBR·Cl**	-1.36	-0.80	-0.49	-0.40	-0.23	0.35
	Physical Mixture*	-1.46	-0.71	-0.46	0.01	0.31	0.66
	HTD1801	-0.74	-0.23	0.07	0.26	0.50	0.70
UDCA	**UDCA**	>5.00	>5.00	2.85	1.37	1.01	1.00
	Physical Mixture*	>5.00	>5.00	2.83	1.46	1.11	1.07
	HTD1801	>5.00	>5.00	>5.00	1.16	1.61	1.06

*1:1 physical mixture of BBR·Cl and UDCA.

- Lipophilicity was substantially enhanced across the pH range for BBR and at pH 6.5 for UDCA as HTD1801 vs BBR·Cl or UDCA alone

Improved Dissolution Profiles of BBR and UDCA from HTD1801

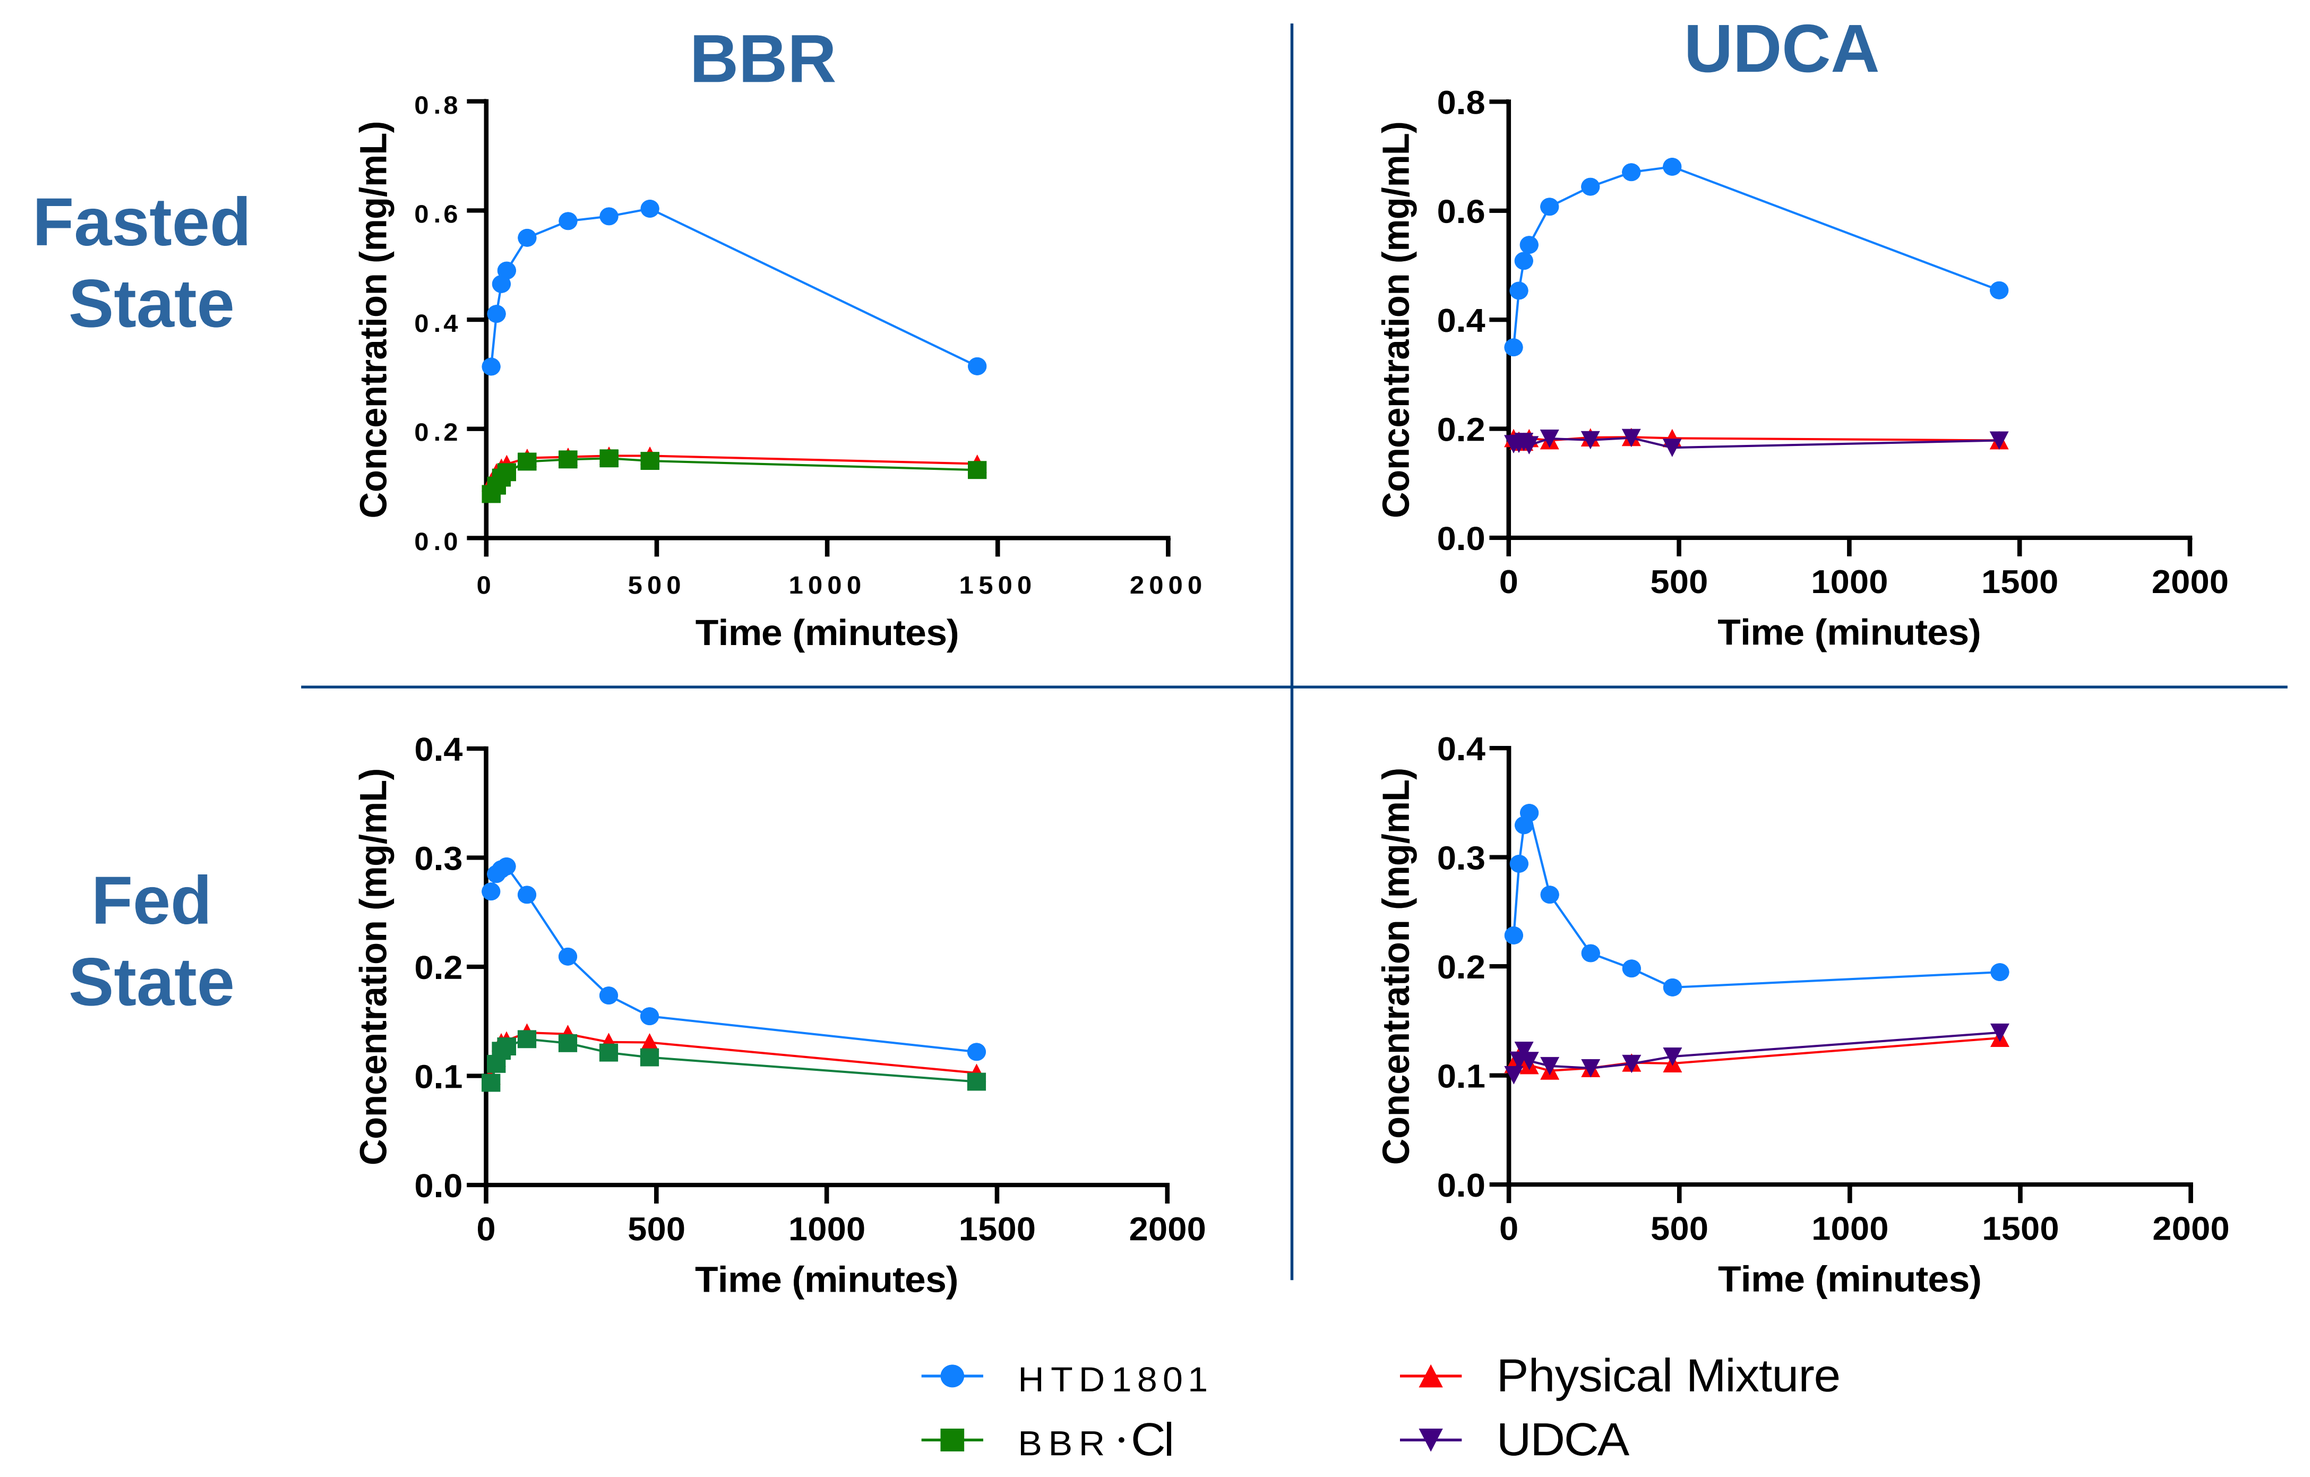
BBR
UDCA
Fasted State
Fed State
Concentration (mg/mL)
Time (minutes)
HTD1801
BBR·Cl
Physical Mixture
UDCA

- Dissolution of BBR and UDCA from HTD1801 showed peak concentrations:
 - ~ >3-fold vs BBR·Cl and UDCA alone in fasted state media
 - ~ >2-fold vs BBR·Cl and UDCA alone in fed state media
- Dissolution of BBR and UDCA as a physical mixture behaved similarly to BBR·Cl and UDCA alone

Significant Improvement in Liver Biochemistry and Lipids with HTD1801 in a Hamster Model of NAFLD

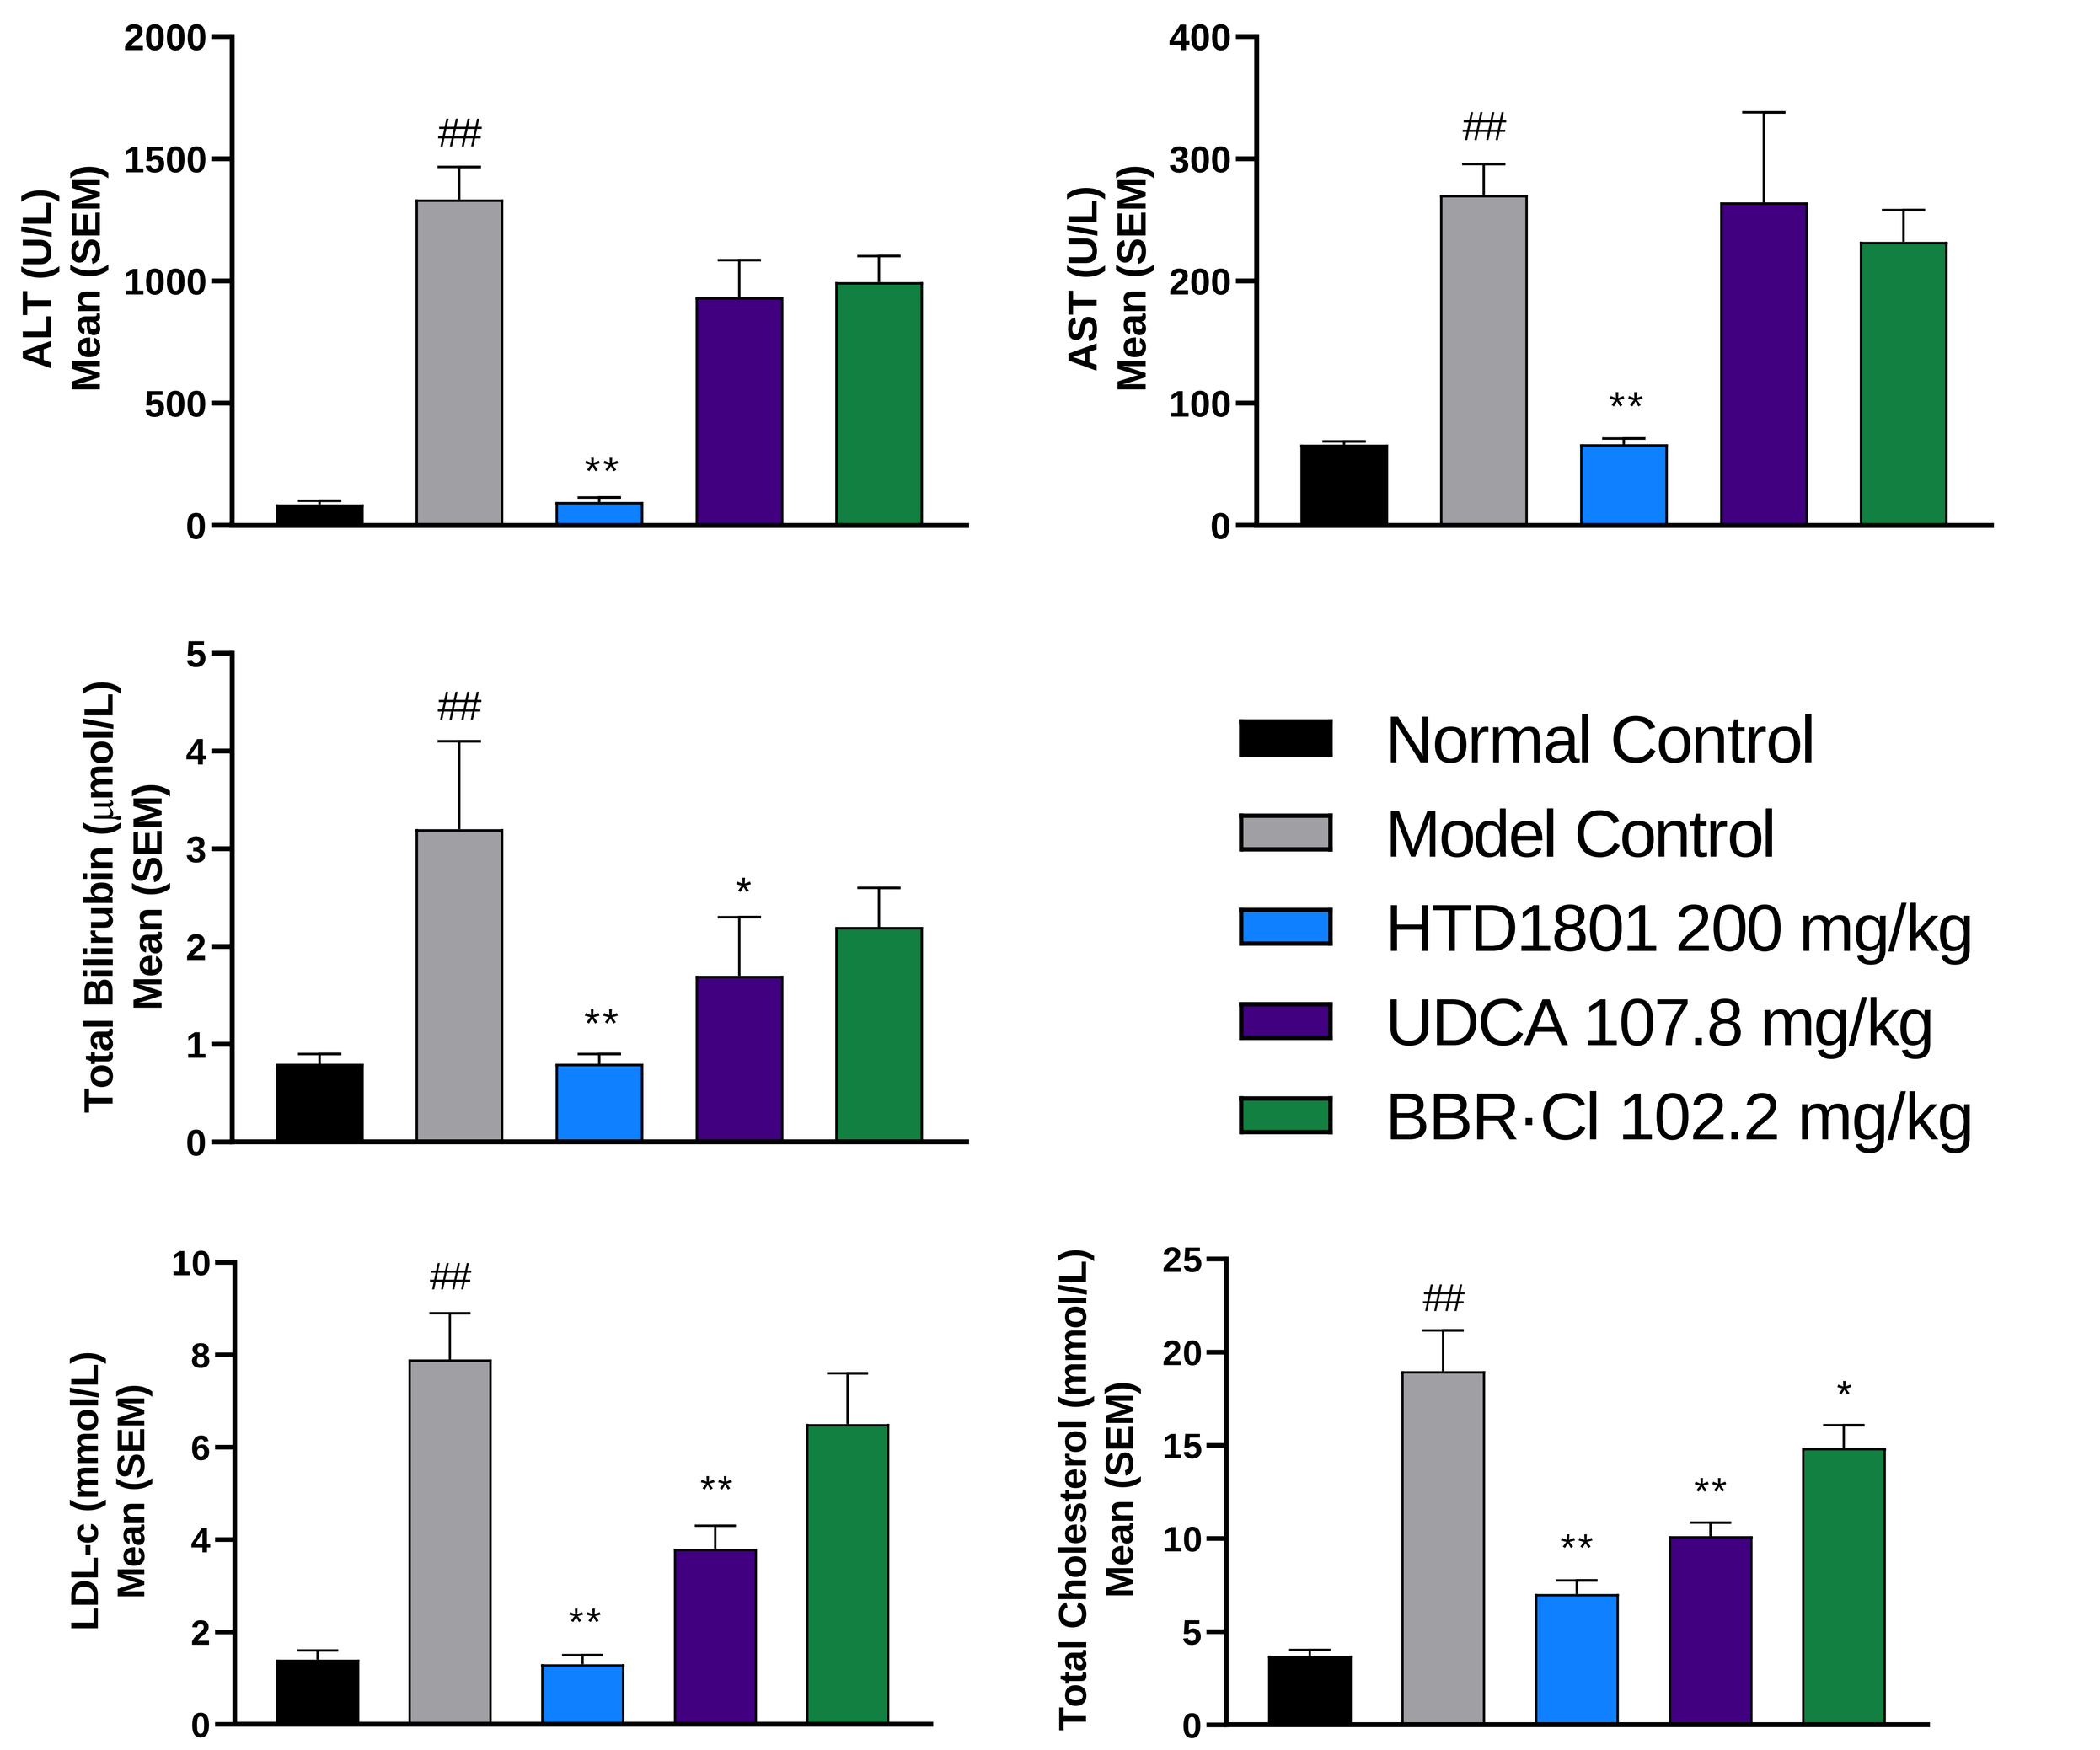
ALT (U/L) Mean (SEM)
AST (U/L) Mean (SEM)
Total Bilirubin (μmol/L) Mean (SEM)
LDL-c (mmol/L) Mean (SEM)
Total Cholesterol (mmol/L) Mean (SEM)
Normal Control
Model Control
HTD1801 200 mg/kg
UDCA 107.8 mg/kg
BBR·Cl 102.2 mg/kg

#P<0.05, ##P<0.01 vs the normal control group.
*P<0.05, **P<0.01 vs the model control group.
Note: Outliers were determined using the Grubbs test and excluded from the analysis.

Significant Histologic and Visual Improvements with HTD1801 in a Hamster Model of NAFLD

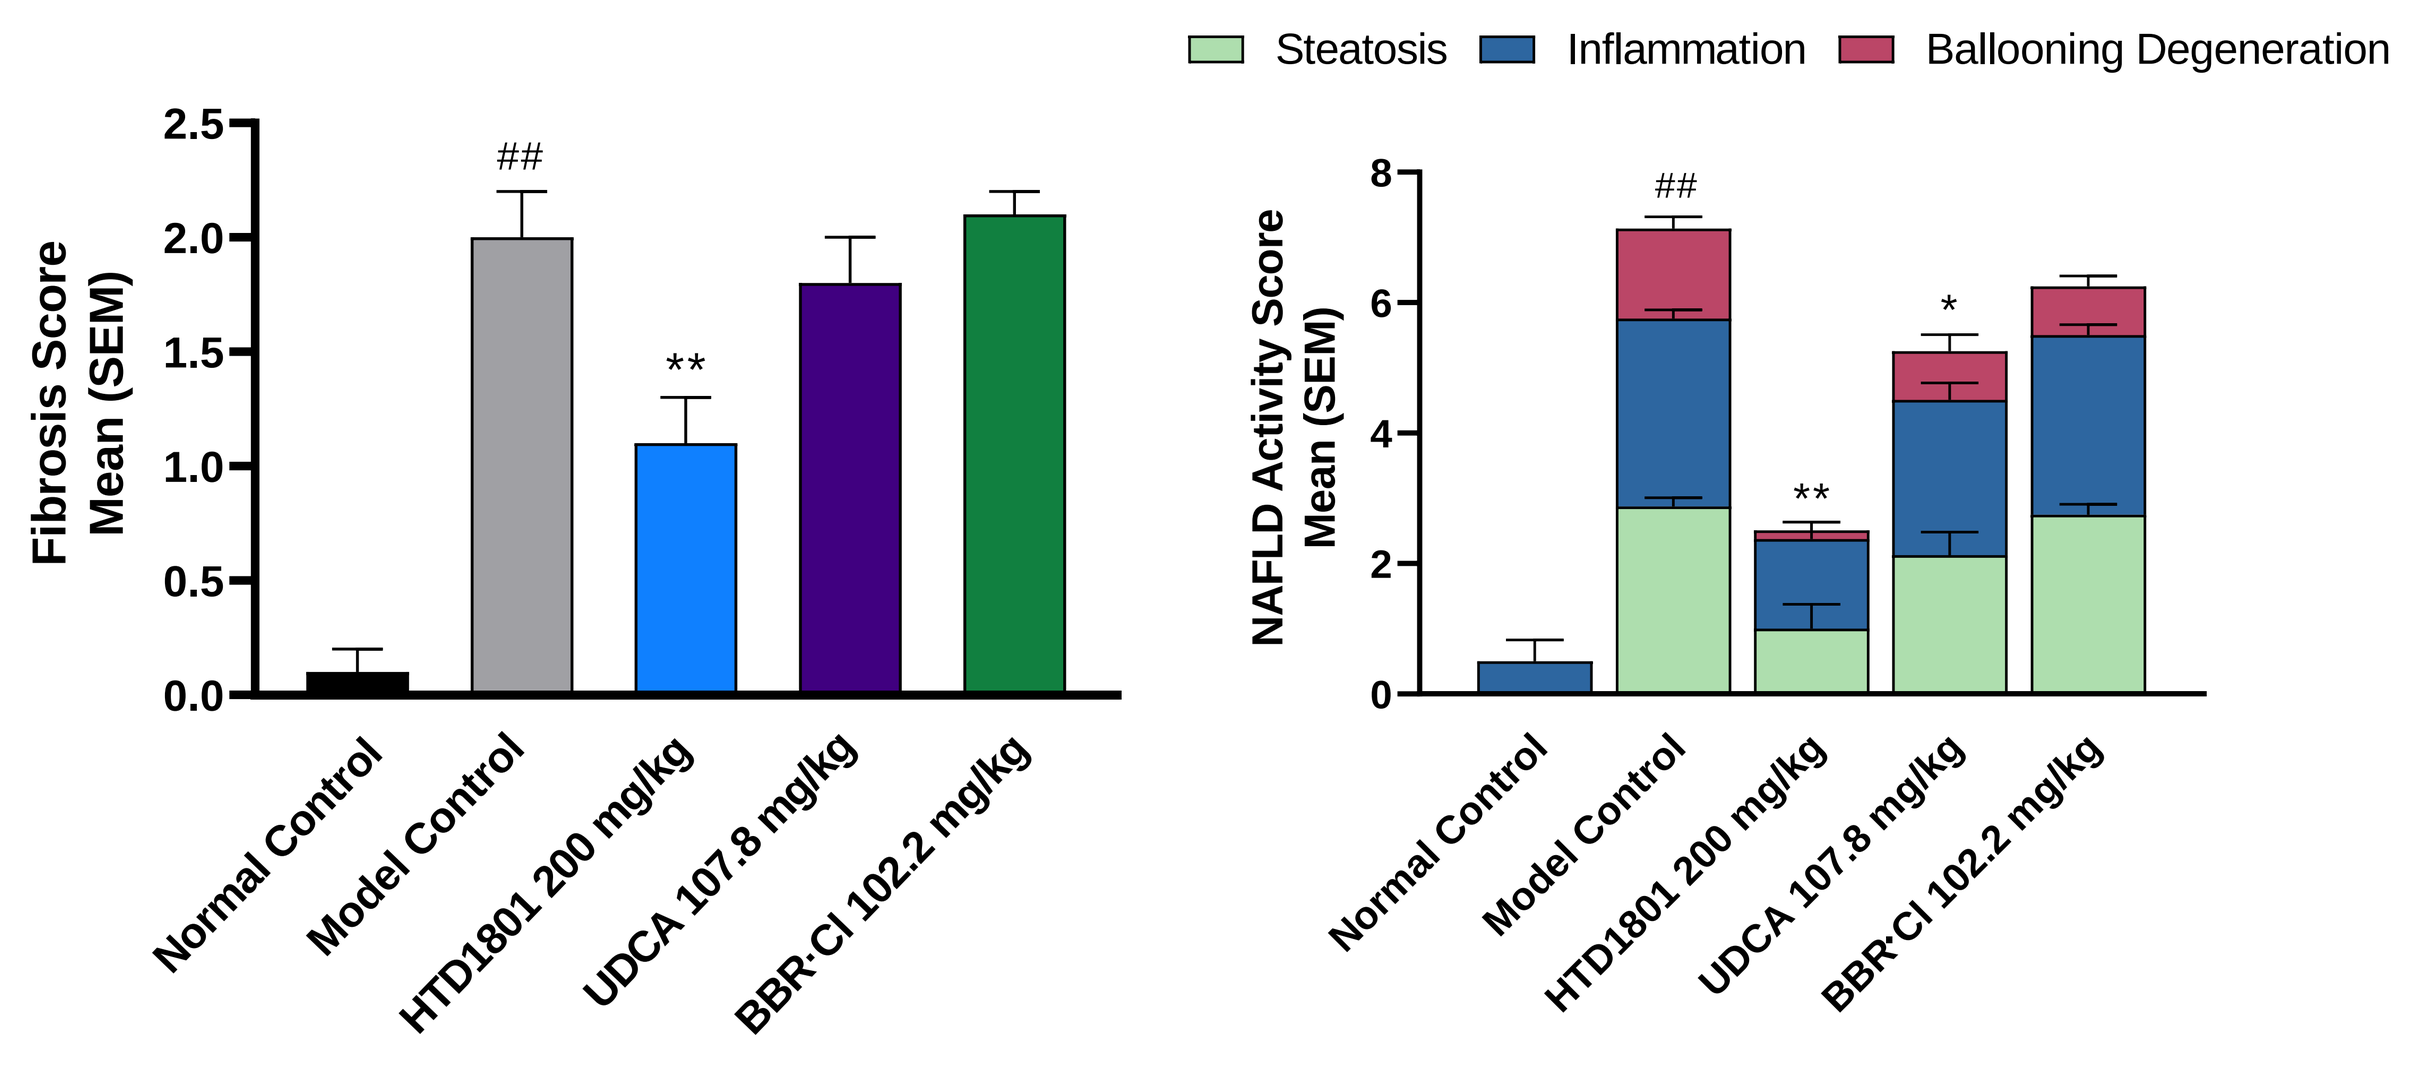
Steatosis
Inflammation
Ballooning Degeneration
Fibrosis Score Mean (SEM)
NAFLD Activity Score Mean (SEM)
Normal Control
Model Control
HTD1801 200 mg/kg
UDCA 107.8 mg/kg
BBR·Cl 102.2 mg/kg

#P<0.05, ##P<0.01 vs the normal control group.
*P<0.05, **P<0.01 vs the model control group.
Note: Outliers were included in the analysis.

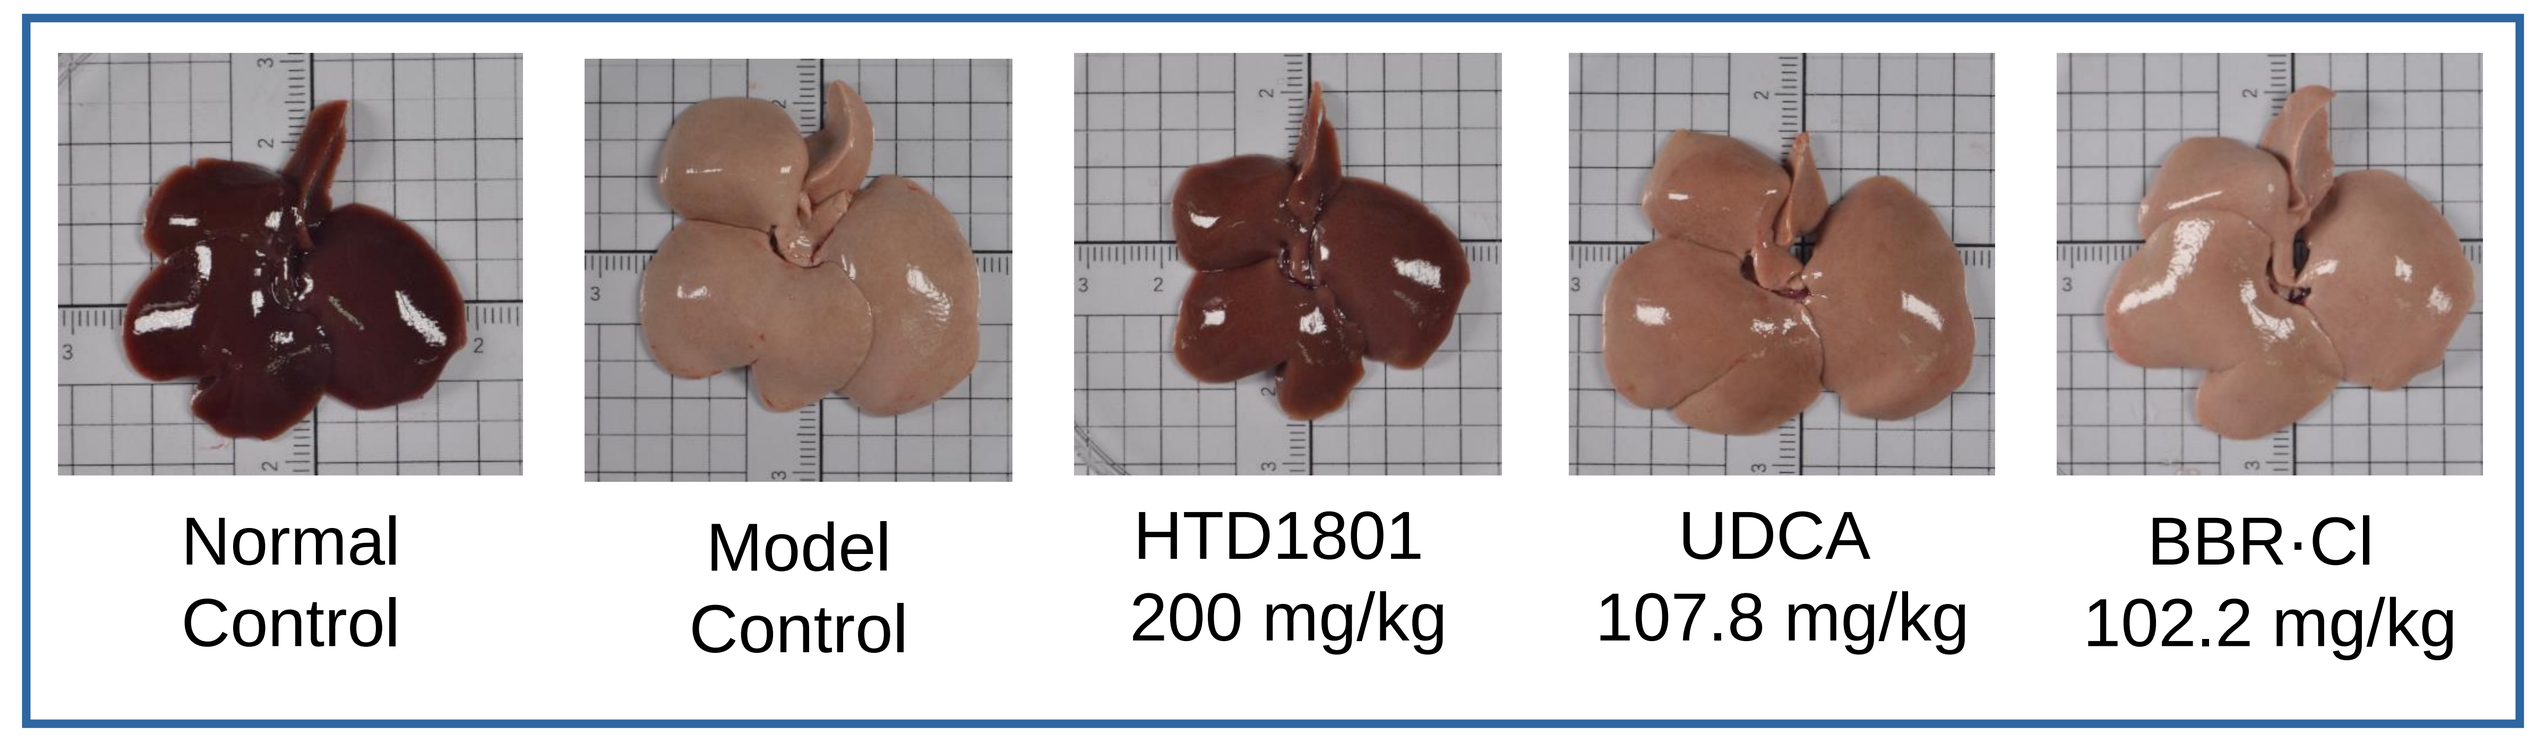
Normal Control
Model Control
HTD1801 200 mg/kg
UDCA 107.8 mg/kg
BBR·Cl 102.2 mg/kg

SUMMARY

- The physicochemical properties of HTD1801 are distinct and superior to UDCA and BBR alone or in a physical mixture
- Studies in the NASH hamster model showed that HTD1801:
 - Significantly reduced ALT, AST, total bilirubin, LDL-c and total cholesterol
 - Significantly improved histology including fibrosis and the NAFLD Activity Score
 - In contrast, UDCA or BBR alone generally resulted in minimal changes in biochemical or histologic parameters

Conclusion

These data show that HTD1801 has physicochemical properties distinct from UDCA or BBR that provide improved dissolution and lipophilicity characteristics which may account for the superior effects seen in NASH and possibly hyperlipidemia and PSC

Contact Information: HighTide Therapeutics, 11140 Rockville Pike, Suite 100-551, Rockville, MD 20852-3149 USA info@hightidetx.com